MON-LB092

Understanding the Patient-Centered Impact of Hypoparathyroidism on Functioning and Well-Being

Meryl Brod,[1] Laura Tesler Waldman,[1] Alden Smith,[2] David B. Karpf,[2]
[1]The Brod Group, [2]Ascendis Pharma, Inc.

BACKGROUND

- Hypoparathyroidism (HP) is a rare endocrine disorder characterized by absent or inappropriately low levels of circulating parathyroid hormone (PTH)[1,2]
- Patients with HP on standard-of-care and/or short-acting PTH therapy report debilitating physical and cognitive symptoms, which may be indicative of a reduced quality of life (QOL)[3-5]
- However, a paucity of data exists on the short-term symptoms and burden of illness experienced by patients with this condition

OBJECTIVE

To investigate the symptoms experienced by adult patients with HP and the impacts of HP on patient QOL.

METHODS

- Using a patient-centered, qualitative study design, semi-structured individual interviews were conducted by telephone with 42 adult patients with post-surgical or idiopathic HP
- Inclusion criteria:
 - At least 18 years old
 - Ability to read, write, and speak English
 - A diagnosis of HP of either at least 6 months (post-surgical), or 12 months (auto-immune or idiopathic)
 - Stable (defined as infrequent severe hypo- or hypercalcemia with high–normal to elevated urine calcium excretion ≥200/250 mg for adults on thiazides or ≥225/275 mg not on thiazides) on standard-of-care (oral calcium and vitamin D supplements) and/or short-acting PTH replacement therapy for at least 3 months
- Exclusion criteria:
 - HP due to CASR mutation or pseudo-hypoparathyroidism
 - Having concomitant condition that may affect calcium or phosphate metabolism
 - Impaired renal dysfunction (Chronic Kidney Disease Stage 4 or 5 (eGFR ≤30 ml/min/1.73 m^2)
 - Cognitive impairment or other medical condition, including psychiatric disorders, that would impact one's ability to participate
- Interviews were conducted with a semi-structured interview guide that was developed based on a literature review and preliminary discussions with clinical experts
- Transcripts were coded utilizing qualitative analysis software and analyzed for content by theme using an adapted grounded theory approach common to qualitative research
- An Independent Review Board approved the study

This study was sponsored by Ascendis Pharma Inc.

RESULTS

Participant Sample Description

- The majority of participants were female (n=35, 83%)
- The primary etiology of reported hypoparathyroidism was post-surgical (n=36, 86%)
- Participants reported having an average of 5.0 (range 0-20) comorbidities in addition to HP
- 71% (n=30) rated the difficulty of managing their HP as somewhat, a lot, or extremely difficult
- 64% (n=27) further reported that it was difficult to find primary care physicians, specialists, and emergency department physicians with sufficient knowledge about HP

Demographic and Health Characteristics	Total (n=42)
Gender, n (%)	
Female	35 (83)
Age	
Mean (median, range)	53 (56, 26-76)
Marital status, n (%)	
Married/partnered	27 (64)
Race/ethnicity, n (%)	
White/Caucasian	40 (95)
Household Income, n (%)	
>US $50,000	21 (50)
Decline to answer	11 (26)
Work status, n (%)	
Work full-time or part-time for pay	21 (50)
Retired	11 (26)
Source of hypoparathyroidism, n (%)	
Post-surgical	36 (86)
Idiopathic	6 (14)
Duration of diagnosis (years)	
Mean (median, range)	14 (9, 0.6-49)
Self-reported other medical conditions, n (%)[a]	
Hypothyroidism	30 (71)
Osteoarthritis	16 (38)
Depression	12 (29)
Chronic back pain	10 (24)
Reflux disease	9 (21)
Thyroid cancer	8 (19)
Medications and supplements currently used to treat HP, n (%)[a]	
Calcium supplements	40 (95)
Vitamin D supplements	41 (98)
PTH replacement therapy (PTH [1-84])	22 (52)
Hydrochlorothiazide (HCTZ)	7 (17)
Magnesium supplements	24 (57)
Potassium supplements	7 (17)

[a]Participants could choose more than one response.

Signs and Symptoms of HP

- 100% of the sample reported experiencing physical signs and symptoms that were attributed to HP, including:
 - Paresthesia (n=37, 88%)
 - Muscle cramping (n=36, 86%)
 - Physical fatigue (n=35, 83%)
- 86% (n=36) further reported experiencing one or more forms of cognitive dysfunction, often collectively referred to as "brain fog", which included:
 - Impaired memory (n=24, 57%)
 - Impaired ability to have a conversation/speech issues (n=21, 50%)
 - Lack of concentration/focus (n=18, 43%)

Most Frequently Reported Signs and Symptoms, n (%)	Total (n=42)
Physical Signs and Symptoms	
Tingling/numbness/paresthesia	37 (88)
Tingling without numbness	20 (48)
Tingling with numbness	15 (36)
Unspecified	2 (5)
Muscle cramping	36 (86)
Fatigue	35 (83)
Tiredness	29 (69)
Less/low energy	28 (67)
Exhaustion	9 (21)
Flu-like symptoms	3 (7)
Lethargy	2 (5)
Pain	32 (74)
Sleep issues	21 (50)
Muscle spasms/twitching	20 (48)
Spasms	14 (33)
Twitching	8 (19)
Both	3 (7)
Muscle weakness/fatigue/stamina	17 (40)
Heat intolerance	14 (33)
Cardiovascular symptoms	14 (33)
Cognitive Signs and Symptoms	
Brain fog	32 (76)
Memory/forgetfulness	24 (57)
Short-term memory	20 (48)
Long-term memory	7 (17)
Unspecified if long- or short-term memory	1 (2)
Ability to have a conversation/speech issues	21 (50)
Trouble finding the correct words to say	20 (48)
Other impairments to ability to have a conversation	3 (7)
Lack of concentration/focus	18 (43)
Can't think right/clearly/fuzzy thinking	13 (31)
Understanding information	11 (26)

Impacts of HP on QOL

- 100% reported interference with daily life, including:
 - Ability to do things around the home (n=33, 79%)
 - Overall productivity (n=28, 67%)
- 100% reported psychological impacts, including feeling:
 - Anxious (n=34, 81%)
 - Frustrated (n=27, 64%)
- 95% of participants (n=40) reported impacts on physical functioning, including:
 - Impaired ability to exercise (n=32, 76%)
 - Impaired mobility (n=21, 50%)
- 95% (n=40) reported social impacts, including:
 - Ability to participate in social activities (n=33, 79%)
 - Relationships (n=32, 76%)

Most Frequently Reported Impacts, n (%)	Total (n=42)
Physical Functioning	
Ability to exercise	32 (76)
Impaired mobility	21 (50)
Less physically active than used to be/wants to be	22 (52)
Need a long time to recover after activities	16 (38)
Daily Life	
Ability to do things around the home	33 (79)
Unable to do as much as used to/wants to	28 (67)
Interference with travel	27 (64)
Need to take breaks/pace yourself when doing activities	26 (62)
Interference with work productivity	18 (43)
Need to stop what you're doing due to symptom onset	16 (38)
Ability to complete complex/detail-oriented tasks	15 (36)
Need to plan day around symptoms	15 (36)
Ability to do leisure activities/hobbies	14 (33)
Daily life disrupted by medical visits/hospitalizations	13 (31)
Ability to do errands	13 (31)
Psychological	
Anxious/anxiety	34 (81)
Frustrated	27 (64)
Depressed/sad	26 (62)
Isolated	18 (43)
Reduced self-confidence/self-image	19 (45)
Irritable/short-tempered	18 (43)
Worry	17 (40)
Anger	14 (33)
Social	
Ability to participate in social activities	33 (79)
Relationships	32 (76)
Relationships with family	18 (43)
Relationships with friends	16 (38)
Relationships with spouse/partner	9 (21)
Limited in types of social activities one can participate in	24 (57)
Social withdrawal	22 (52)

Selected Quotes About Signs and Symptoms of HP

"My hands literally cramp up to—they kind of curl up, and into a big cramp, you can't undo them, you can't use your hands, and it's painful."

PHYSICAL: Muscle cramping

"I'll go trying to get something done, and I've got to go to the other room and grab something, and I'll have that brain fog and I'll forget what I went to go grab. Then I'll forget what I was doing."

COGNITIVE: Trouble remembering

Selected Quotes About Impacts of HP on QOL

"It's very difficult and painful, and dangerous, actually, to walk when my legs are numb and tingly."

PHYSICAL FUNCTIONING: Mobility

"It's kind of hard to do dishes when your hands are tingling and you can't feel them, or they're cramping up because you're trying to grab a plate and then you just end up dropping them."

DAILY LIFE: Ability to do things around the home

"Always feeling anxious about doing anything because I don't know how my body is going to deal with it that day."

PSYCHOLOGICAL: Anxious

"When you are not able to go out and participate in all the activities that you used to be able to do, then people forget about you."

SOCIAL: Relationships with friends

LIMITATIONS

- The patients who responded to recruitment outreach were primarily female with post-surgical HP. However, the proportions in this study are similar to those reported in other studies[6,7]
- This study did not focus on the long-term medical complications of HP; further investigation of long-term complications is needed to more completely under stand both the short- and long-term impact of HP

CONCLUSIONS

- Study findings show that almost all HP patients interviewed experienced substantial illness burden, even on treatment, with respect to both the range and severity of symptoms and QOL impacts
- A better understanding of this burden is important to inform clinician treatment decisions and address unmet treatment needs

REFERENCES:
[1]Mannstadt, M., et al. (2017). Nat Rev Dis Primers, 3, p. 17080. [2]Bilezikian, J. P., et al. (2011). J Bone Miner Res, 26(10), p. 2317-2337. [3]Vokes, T. (2019). Bone, 120, p. 542-547. [4]Astor, M. C., et al. (2016). J Clin Endocrinol Metab, 101(8), p. 3045-3053. [5]Cusano, N. E., et al. (2013). J Clin Endocrinol Metab, 98(6), p. 2356-2361. [6]Sikjaer, T., et al. (2014). Osteoporos Int, 25(6), p. 1717-1726. [7]Rubin, M. R., et al. (2016). J Clin Endocrinol Metab, 101(7), p. 2742-2750.

Ascendis®, the Ascendis Pharma logo, the company logo and TransCon® are registered trademarks owned by Ascendis Pharma A/S.
© March 2019 Ascendis Pharma A/S.